POSTER #892

ABL503 (TJ-L14B), PD-L1×4-1BB bispecific antibody, induces superior anti-tumor activity by PD-L1-dependent 4-1BB activation with the increase of 4-1BB⁺CD8⁺ T cells in tumor microenvironment

Gihoon You[1], Uijung Jung[1], Jaehyoung Jeon[1], Shinai Lee[1], Hyung-Seung Jin[2], Youngkwang Kim[1], Eunsil Sung[1], Hyunjoo Kim[1], Yangmi Lim[1], Jonghwa Won[1], Zhengyi Wang[3], Wenqing Jiang[3], Jaeho Jung[1,*]

[1]ABL Bio, INC., Seongnam, Republic of Korea; [2]Asan Medical Center, Seoul, Republic of Korea; [3]I-Mab Biopharma, Shanghai, China

Introduction

PD-1/PD-L1 inhibitor has revolutionized cancer treatment, but there are unmet clinical needs for PD-1/PD-L1 inhibitor-resistant/refractory patients. Activation of T cells in the tumor microenvironment (TME) by 4-1BB agonist antibodies is one of the promising approaches to complement the current limitation of PD-1/PD-L1 inhibitors. Although 4-1BB is a promising target for immunotherapy, clinical studies using 4-1BB agonist antibodies showed systemic immune cell activation resulting in dose-limiting hepatotoxicity. We generated ABL503 (TJ-L14B), a bispecific antibody (BsAb) that combines PD-1/PD-L1 pathway blockade and 4-1BB agonistic activity dependent on the PD-L1 engagement to limit unwanted toxicities while exerting a potent anti-tumor efficacy. Here, we reported the pre-clinical properties of ABL503 (TJ-L14B) in various studies.

Background and Mode of Action

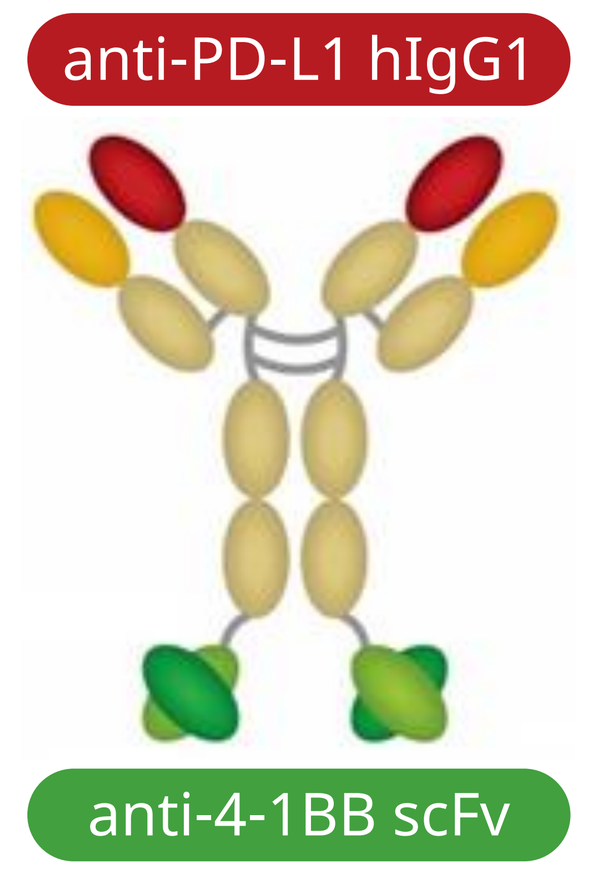
anti-PD-L1 hIgG1
anti-4-1BB scFv

ABL503 (TJ-L14B): Full-length anti-PD-L1 mAb (Fc-silenced human IgG1, hIgG1) fused with scFv of anti-4-1BB mAb

[Functional activity]

1. 4-1BB clustering/activation only in PD-L1 engagement
2. PD-1/PD-L1 pathway inhibition

→ Enhanced anti-tumor activity by simultaneous 4-1BB activation and PD-1/PD-L1 blockade while minimizing the risks of peripheral toxicity

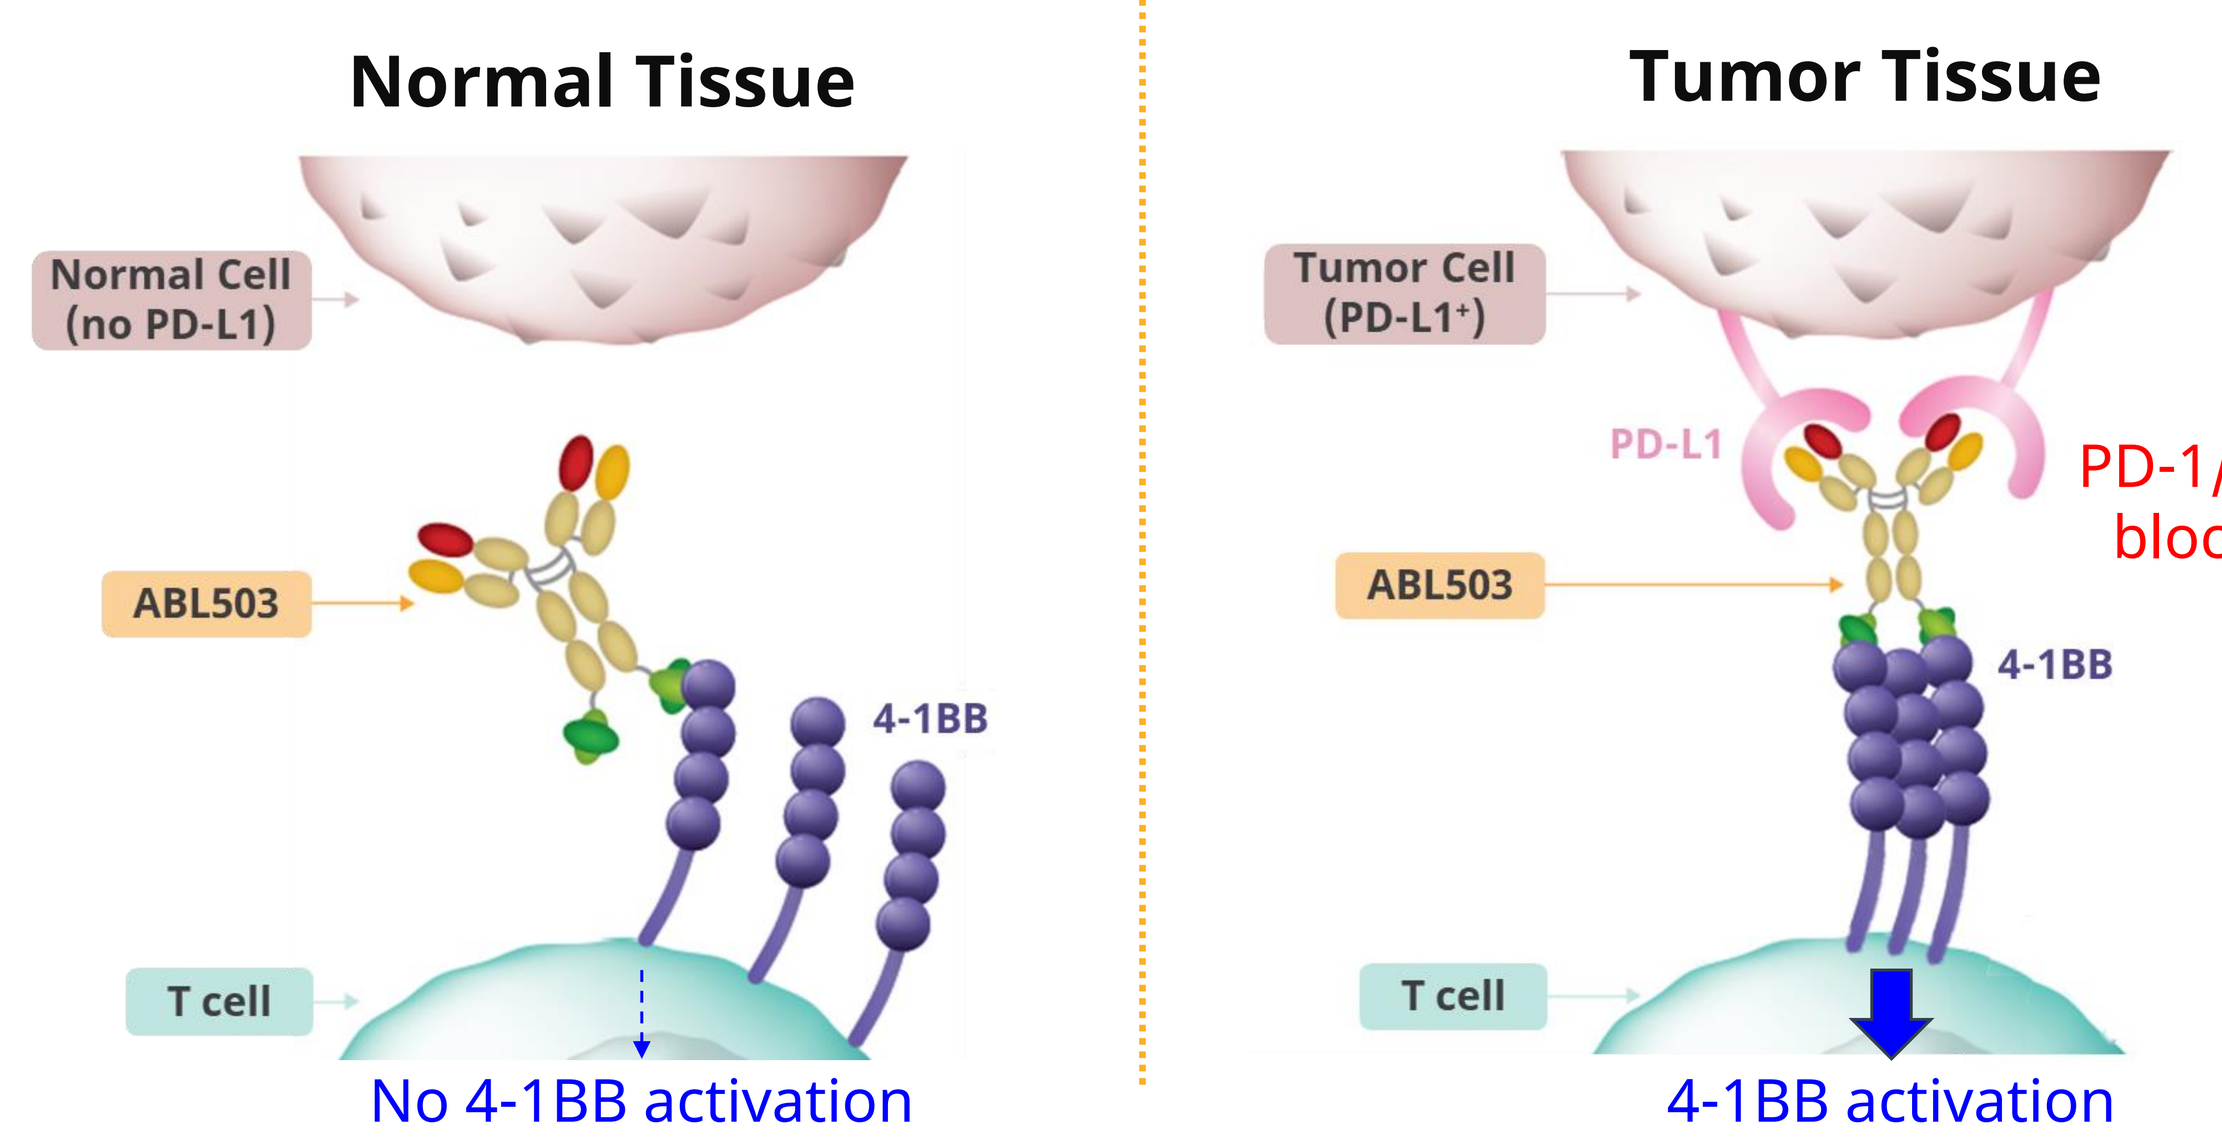
Normal Tissue
Normal Cell (no PD-L1)
ABL503
4-1BB
T cell
No 4-1BB activation
Tumor Tissue
Tumor Cell (PD-L1⁺)
PD-L1
PD-1/PD-L1 blockade
ABL503
4-1BB
T cell
4-1BB activation

Methods

- **PD-L1-dependent 4-1BB agonistic activity** was tested by 4-1BB bioassay with PD-L1-expressing tumor cells and 4-1BB signaling reporter cells. PD-L1 expression was measured by flow cytometry.
- **IFN-γ secretion** was measured in human PBMCs and Calu-6 co-culture system.
- **In vivo efficacy** was evaluated in B6-hPD-L1/h4-1BB knock-in mice implanted with MC38 tumor expressing a different level of hPD-L1.
- **Tumor-infiltrated mCD8⁺ or h4-1BB⁺ cells** were measured by IHC.
- **Pharmacodynamic changes in TILs and blood** were evaluated hPD-L1-expressing MC38 tumor-bearing B6-hPD-L1/h4-1BB knock-in mice.
- **In vitro tumor-killing activity** was assessed using an Autologous organoid-based Discovery for Immuno-Oncology drug platform from ORGANOIDSCIENCE by co-culturing cancer organoids from lung cancer patients with autologous PBMCs.
- **Cytokine (IL-6 and TNF-α) release** was measured in PBMCs from healthy donors stimulated with ABL503 (TJ-L14B).
- Statistical significances were analyzed by *t*-test, one-way ANOVA with Tukey's or Dunnett's multiple comparison test, or two-way ANOVA with Bonferroni's multiple comparison test in GraphPad Prism. *, $p < 0.05$; **, $p < 0.01$; ***, $p < 0.001$; ****, $p < 0.0001$; ns, not significant.

Results

ABL503 shows PD-L1-dependent 4-1BB agonistic activity

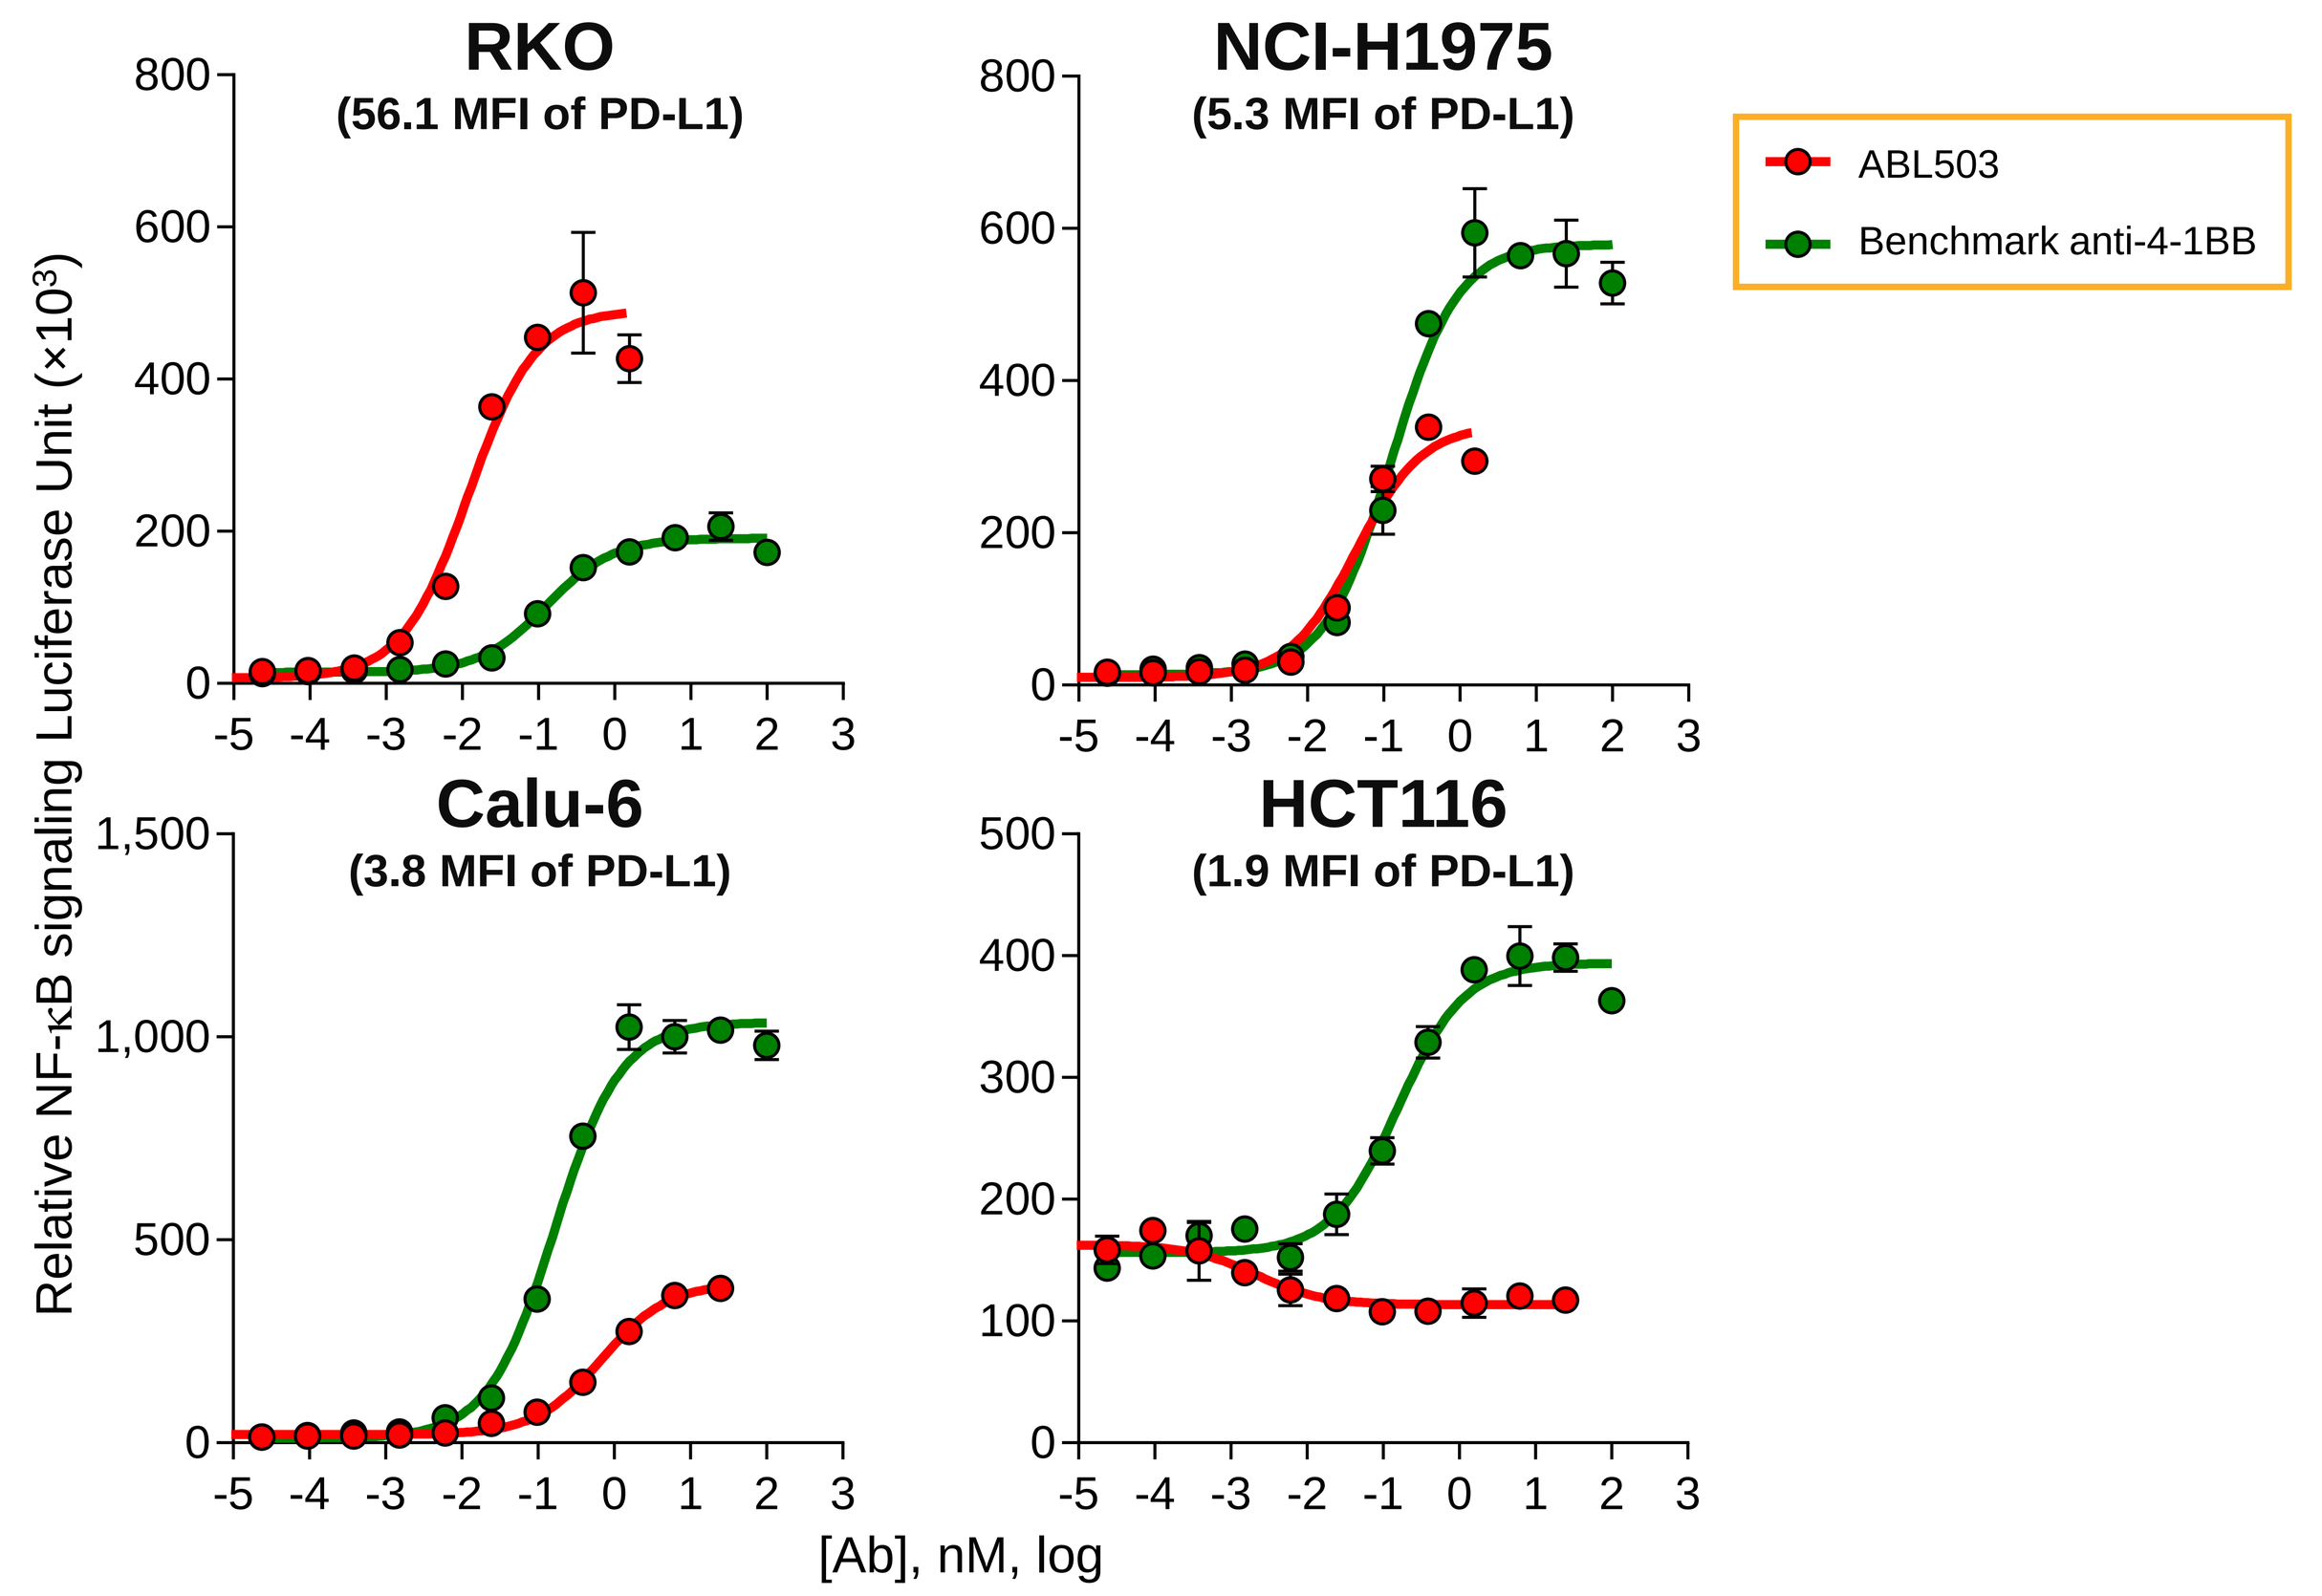
RKO (56.1 MFI of PD-L1)
NCI-H1975 (5.3 MFI of PD-L1)
Calu-6 (3.8 MFI of PD-L1)
HCT116 (1.9 MFI of PD-L1)
ABL503
Benchmark anti-4-1BB
Relative NF-κB signaling Luciferase Unit (×10³)
[Ab], nM, log

- 4-1BB signaling reporter cells and indicated tumor cells were cocultured in the presence of ABL503 or benchmark anti-4-1BB antibody (4-fold dilutions).
- ABL503 (TJ-L14B) showed 4-1BB agonistic activity in a PD-L1-dependent manner, while benchmark anti-4-1BB induced 4-1BB activation regardless of PD-L1.
- Higher PD-L1 expression leads to more potent 4-1BB activation by ABL503 (TJ-L14B).

PD-L1 on immune cells also contribute to the activity of ABL503

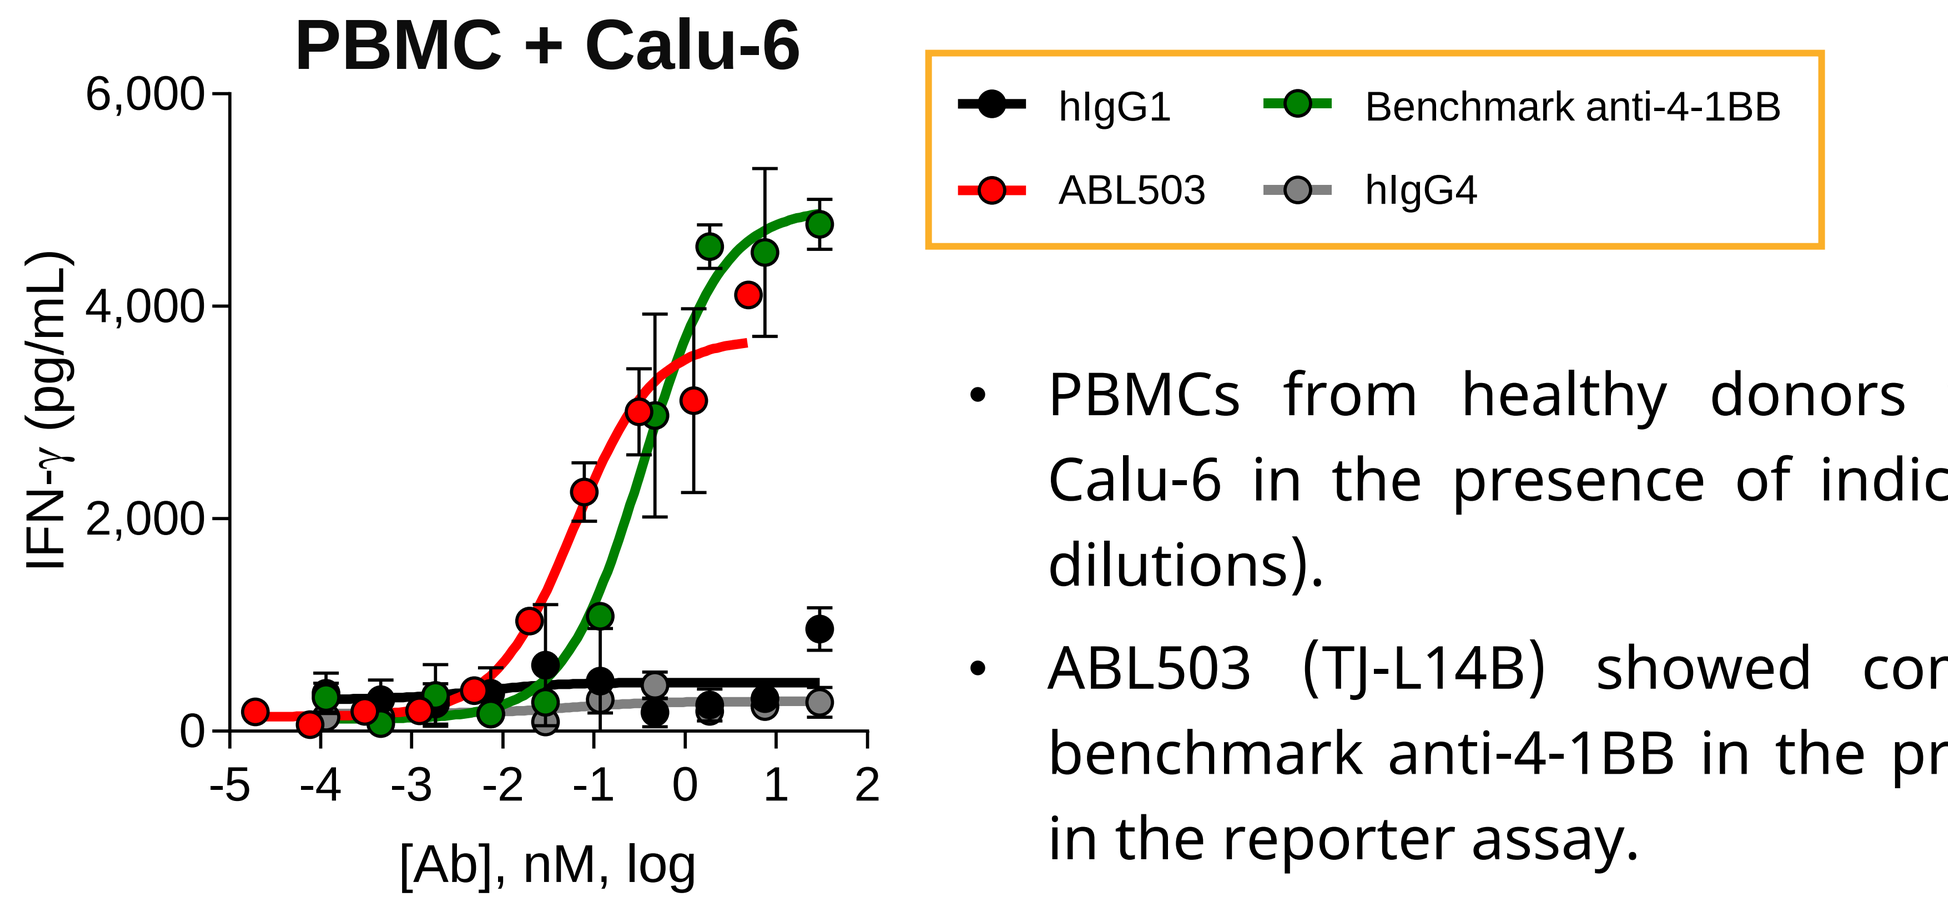
PBMC + Calu-6
hIgG1
ABL503
Benchmark anti-4-1BB
hIgG4
IFN-γ (pg/mL)
[Ab], nM, log

- PBMCs from healthy donors were co-cultured with Calu-6 in the presence of indicated antibodies (4-fold dilutions).
- ABL503 (TJ-L14B) showed comparable activity with benchmark anti-4-1BB in the presence of PBMC unlike in the reporter assay.

ABL503 shows favorable safety profile

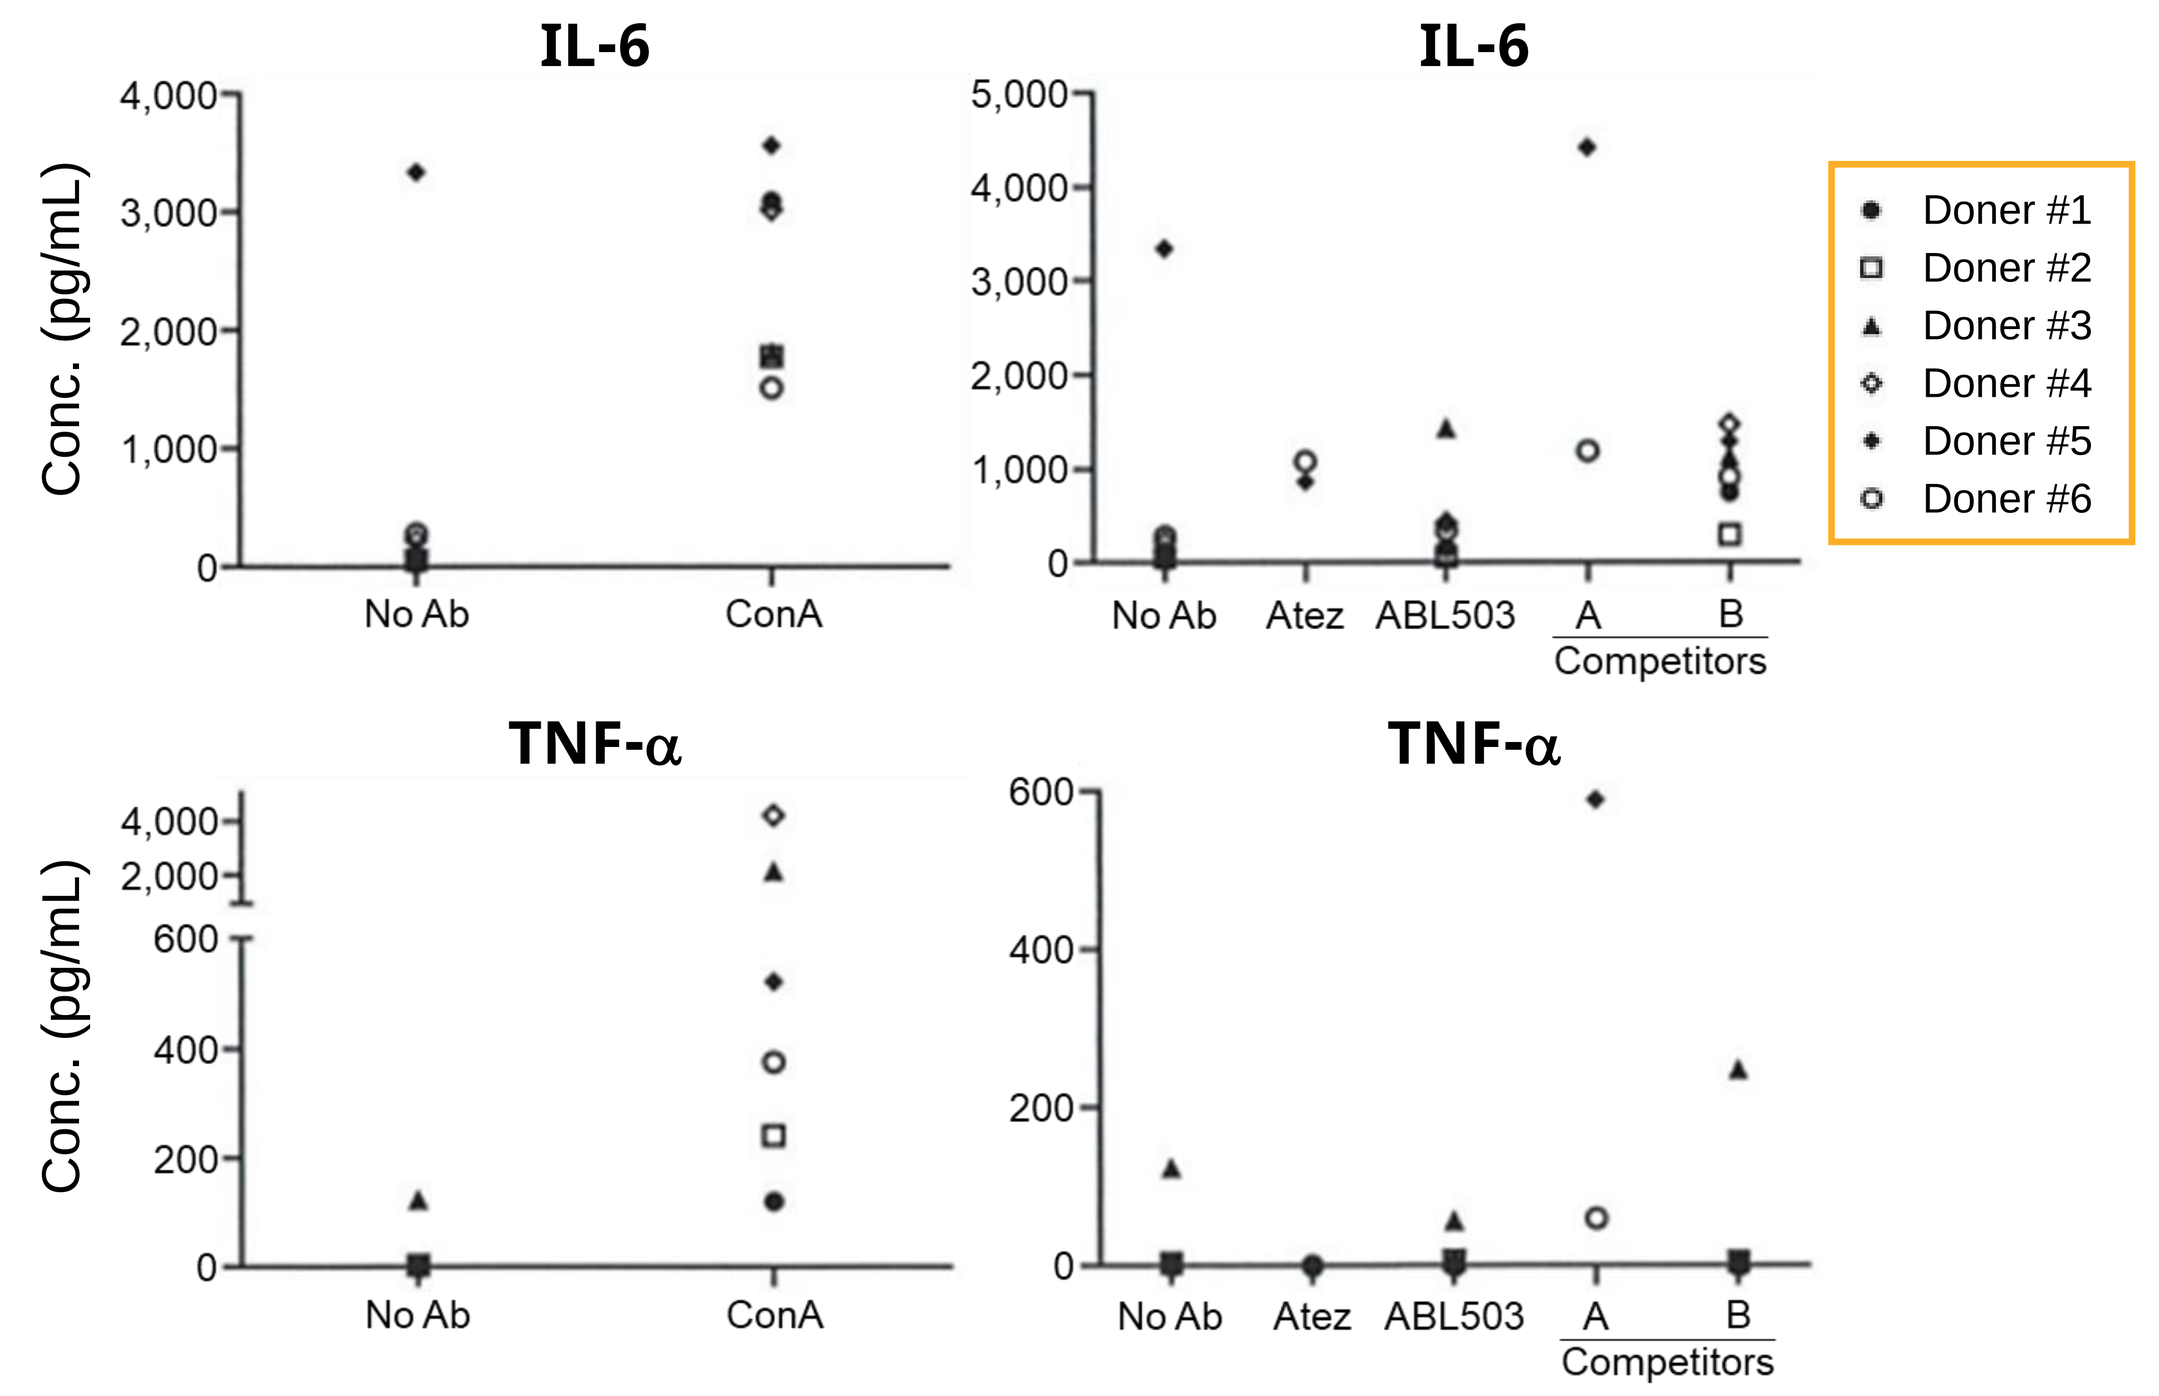
IL-6
TNF-α
Conc. (pg/mL)
No Ab
ConA
Atez
ABL503
A
B
Competitors
Doner #1
Doner #2
Doner #3
Doner #4
Doner #5
Doner #6

- PBMCs from healthy donors were stimulated with indicated antibodies, including Atezolizumab (Atez), ABL503 (TJ-L14B), and two competitor BsAbs (A & B). No Ab control and ConA were used for negative and positive control, respectively. Cytokine release was measured by ELISA.
- No cytokine release by ABL503 (TJ-L14B) was observed.

ABL503 elicits anti-tumor response across different PD-L1 levels

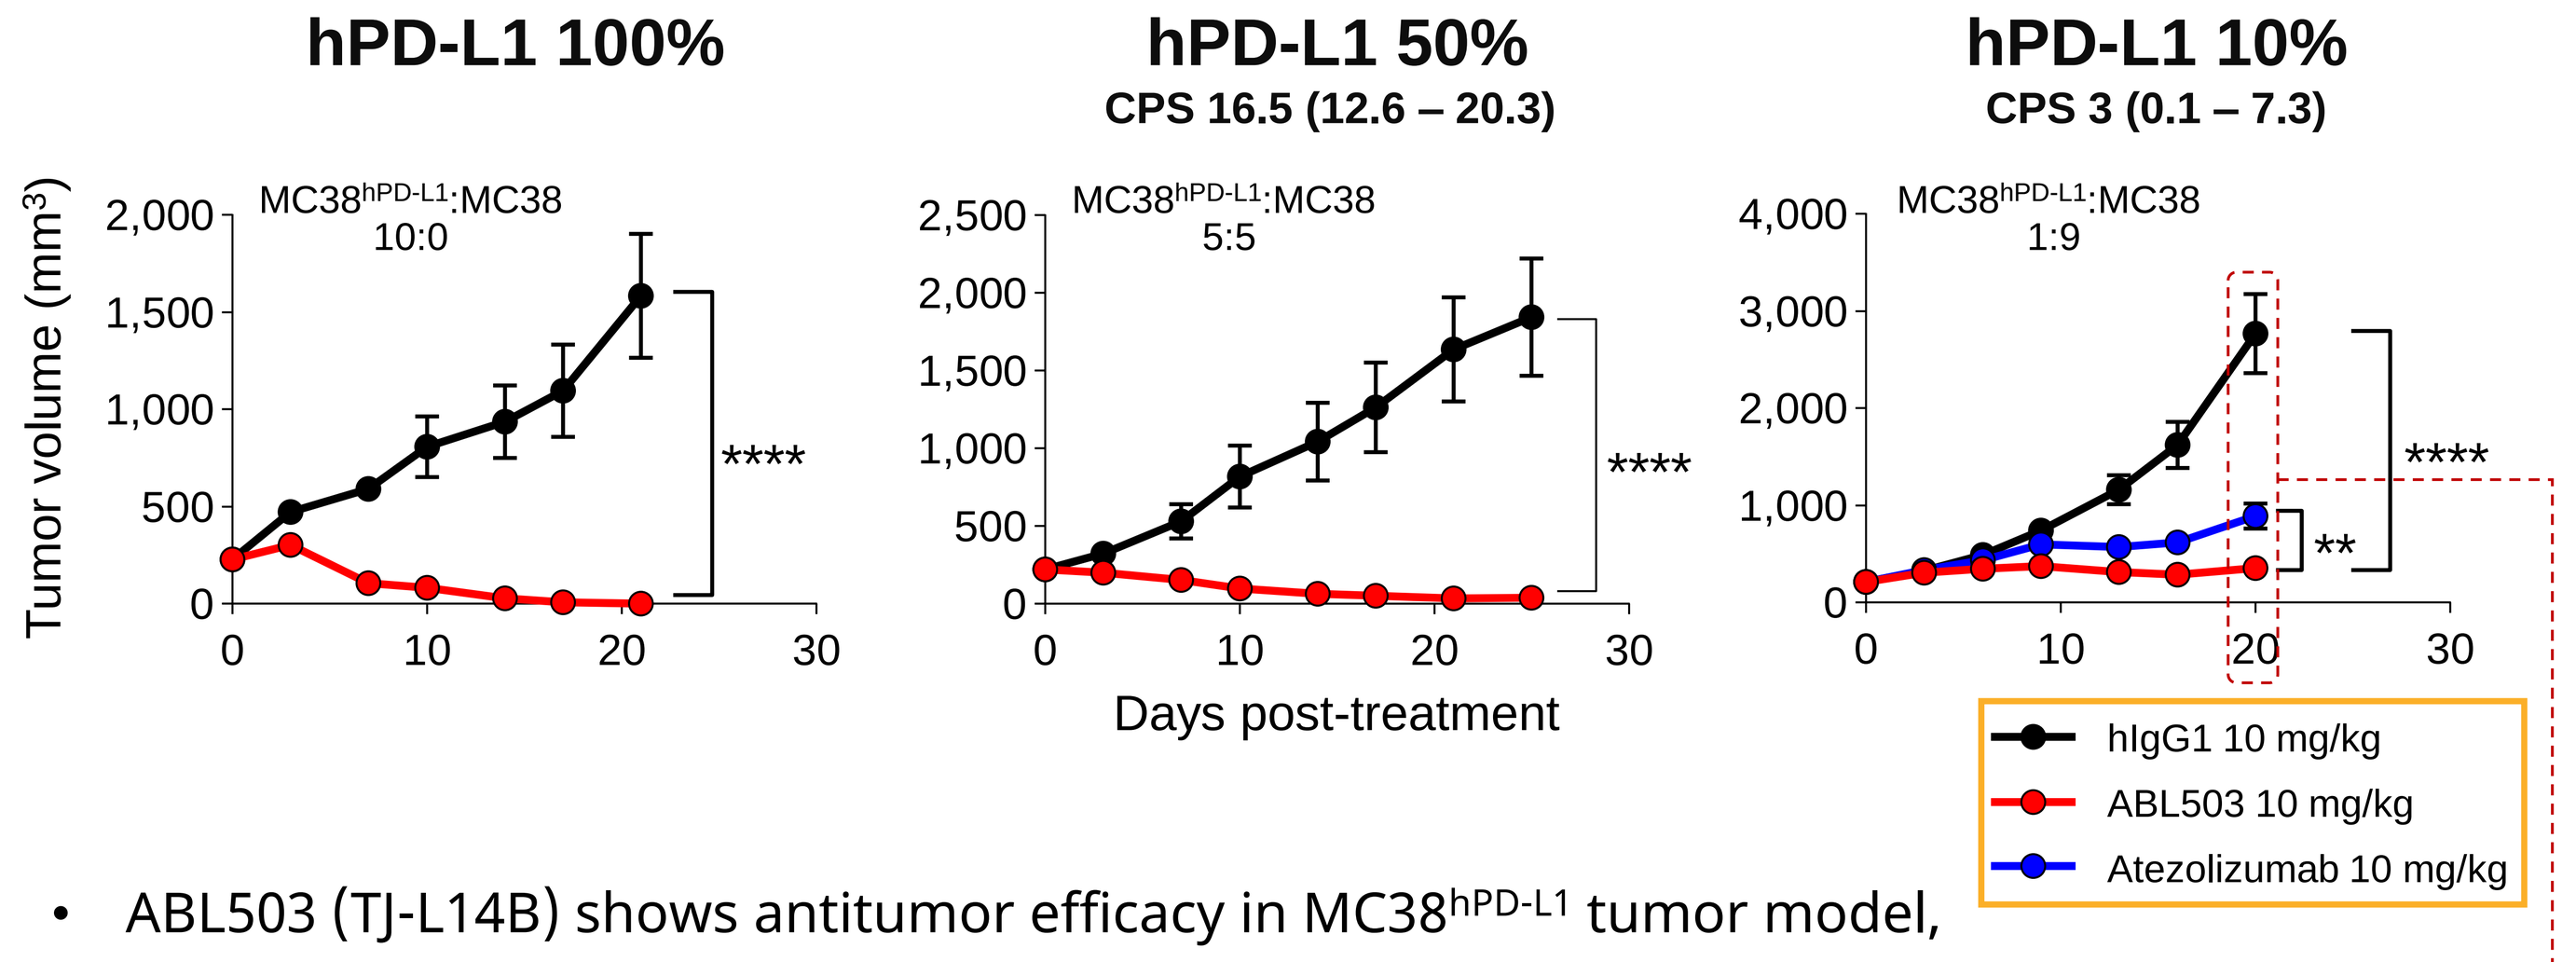
hPD-L1 100%
hPD-L1 50%
CPS 16.5 (12.6 – 20.3)
hPD-L1 10%
CPS 3 (0.1 – 7.3)
Tumor volume (mm³)
Days post-treatment
hIgG1 10 mg/kg
ABL503 10 mg/kg
Atezolizumab 10 mg/kg

- ABL503 (TJ-L14B) shows antitumor efficacy in MC38^{hPD-L1} tumor model, even in the model in which only 10% of tumor cells express hPD-L1 (MC38$^{hPD-L1\ low}$).
- ABL503 (TJ-L14B) shows better activity over Atezolizumab in MC38$^{hPD-L1\ low}$ tumor model.

ABL503 increases CD8⁺ and 4-1BB⁺ cells in the MC38$^{hPD-L1\ low}$ tumor

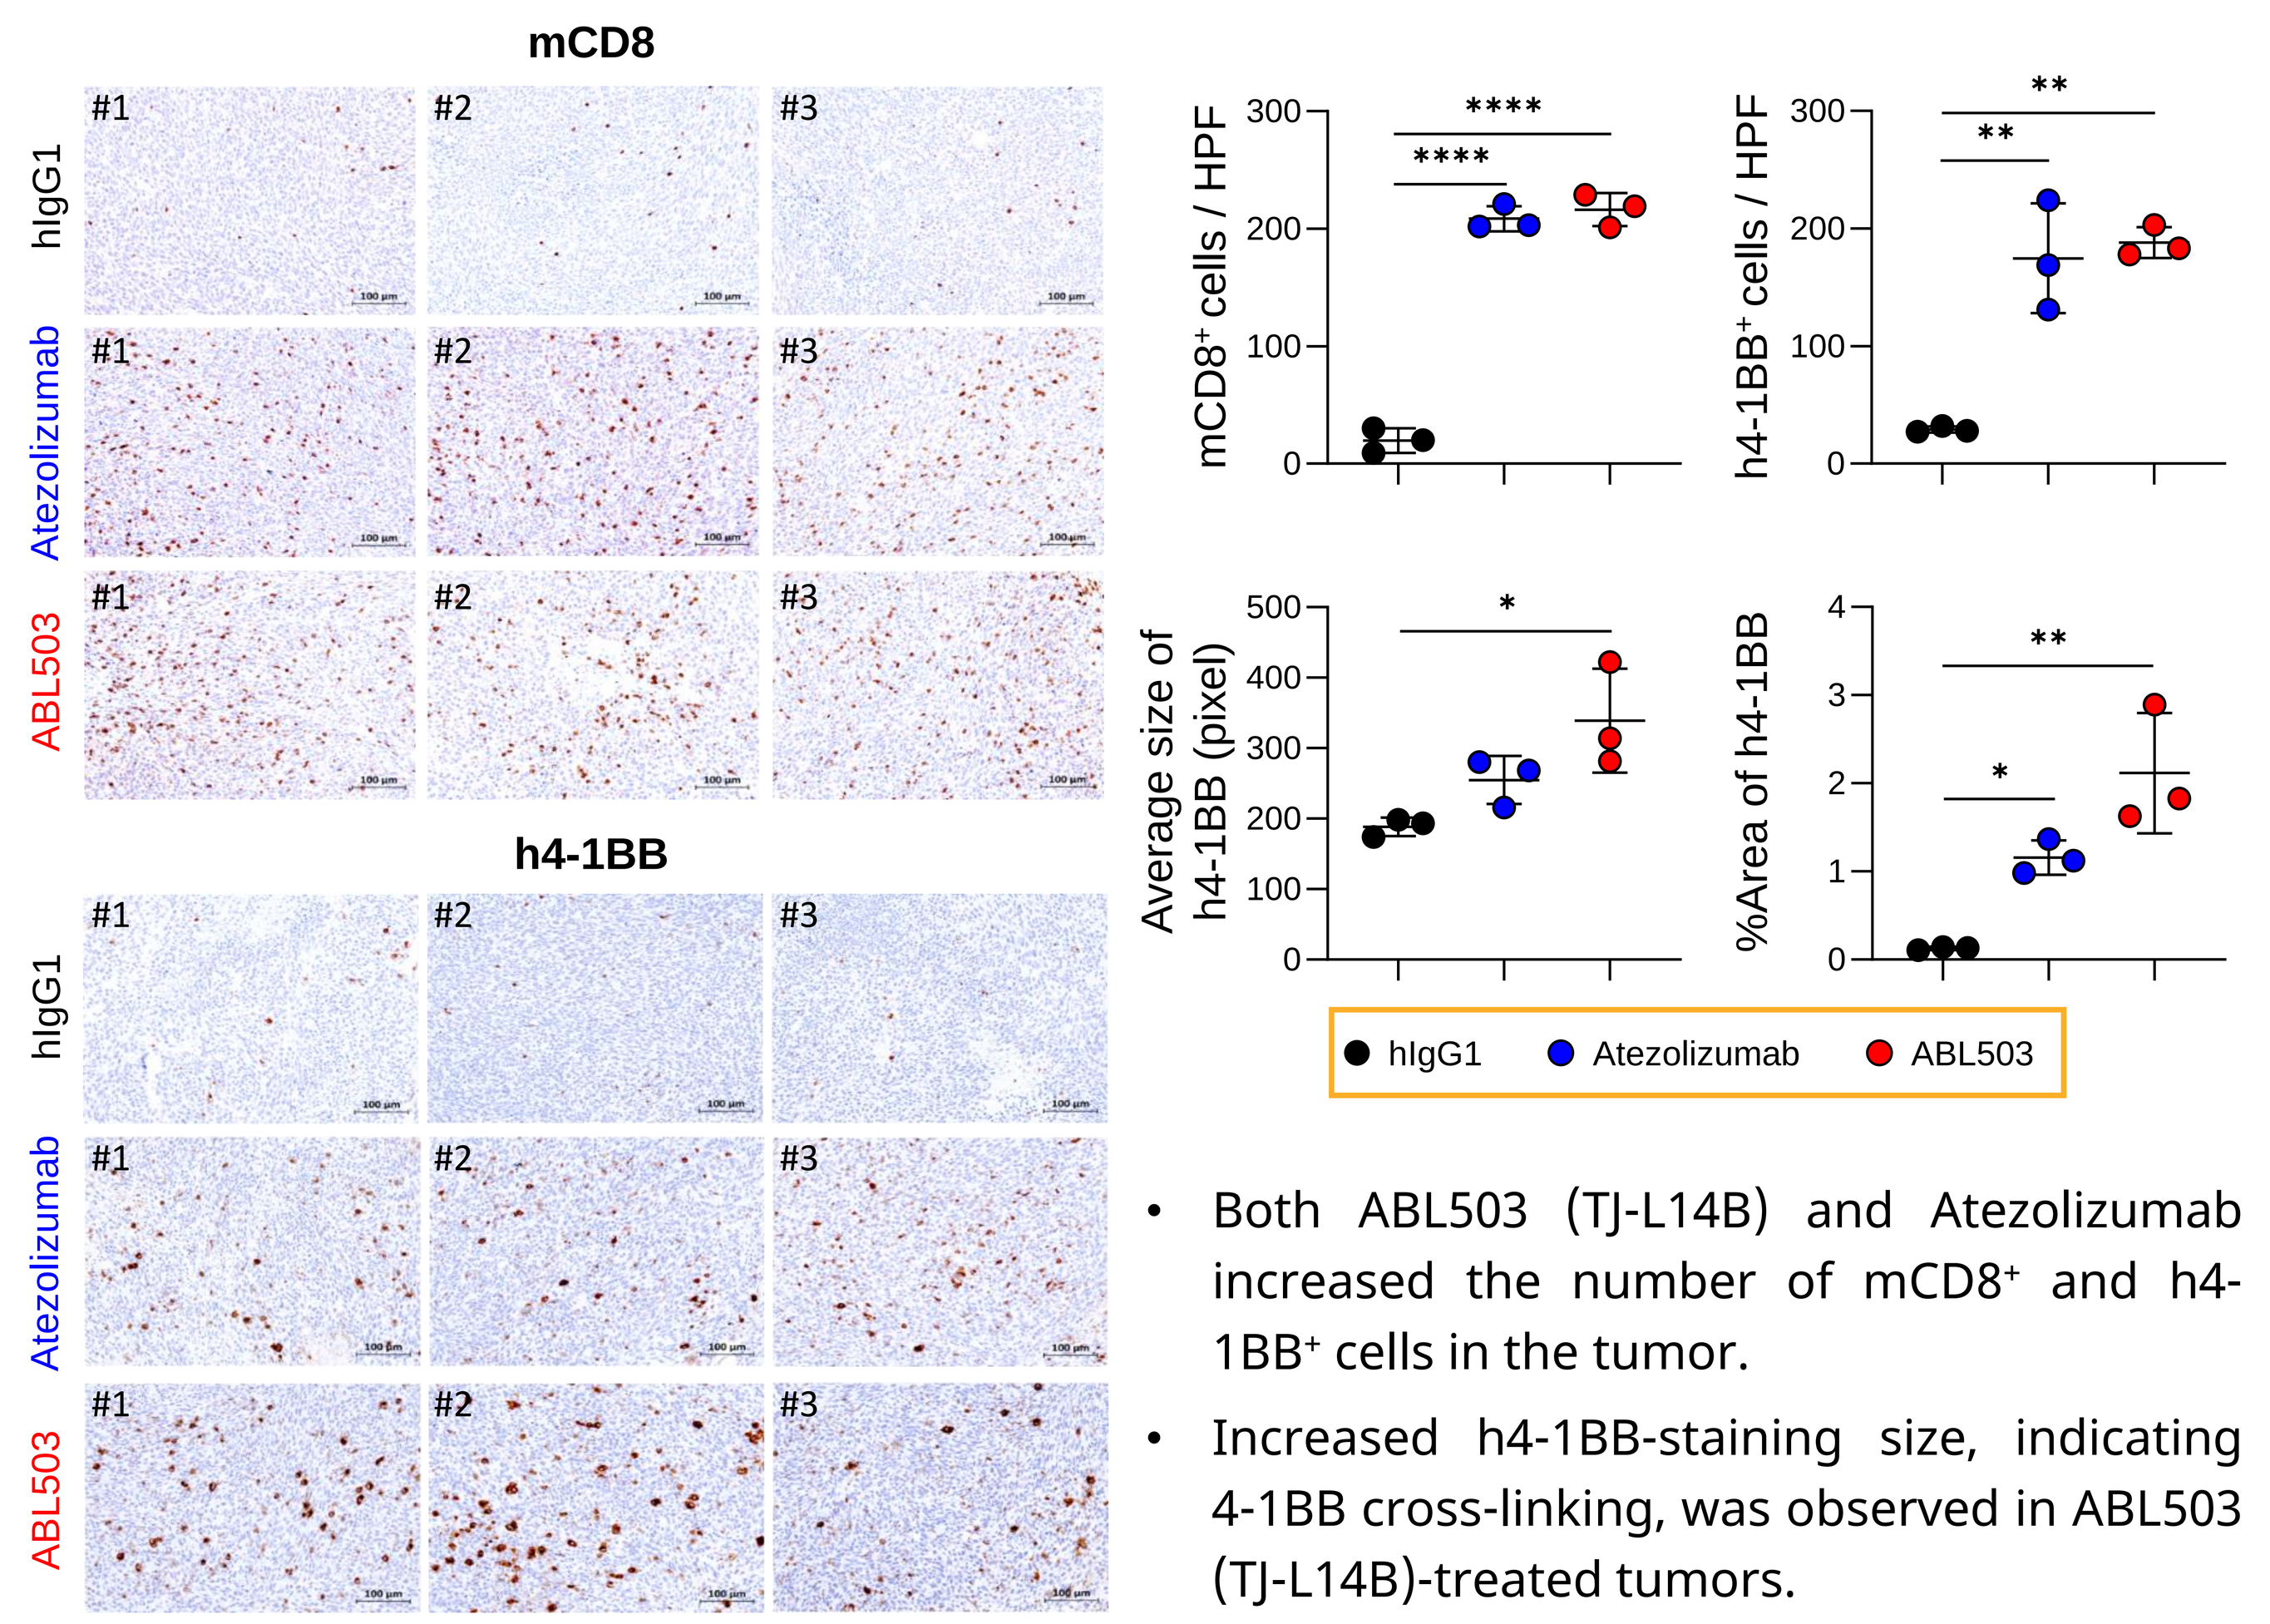
mCD8
h4-1BB
hIgG1
Atezolizumab
ABL503
mCD8⁺ cells / HPF
h4-1BB⁺ cells / HPF
Average size of h4-1BB (pixel)
%Area of h4-1BB

- Both ABL503 (TJ-L14B) and Atezolizumab increased the number of mCD8⁺ and h4-1BB⁺ cells in the tumor.
- Increased h4-1BB-staining size, indicating 4-1BB cross-linking, was observed in ABL503 (TJ-L14B)-treated tumors.

ABL503 shows superior activity over Atezolizumab in organoid system

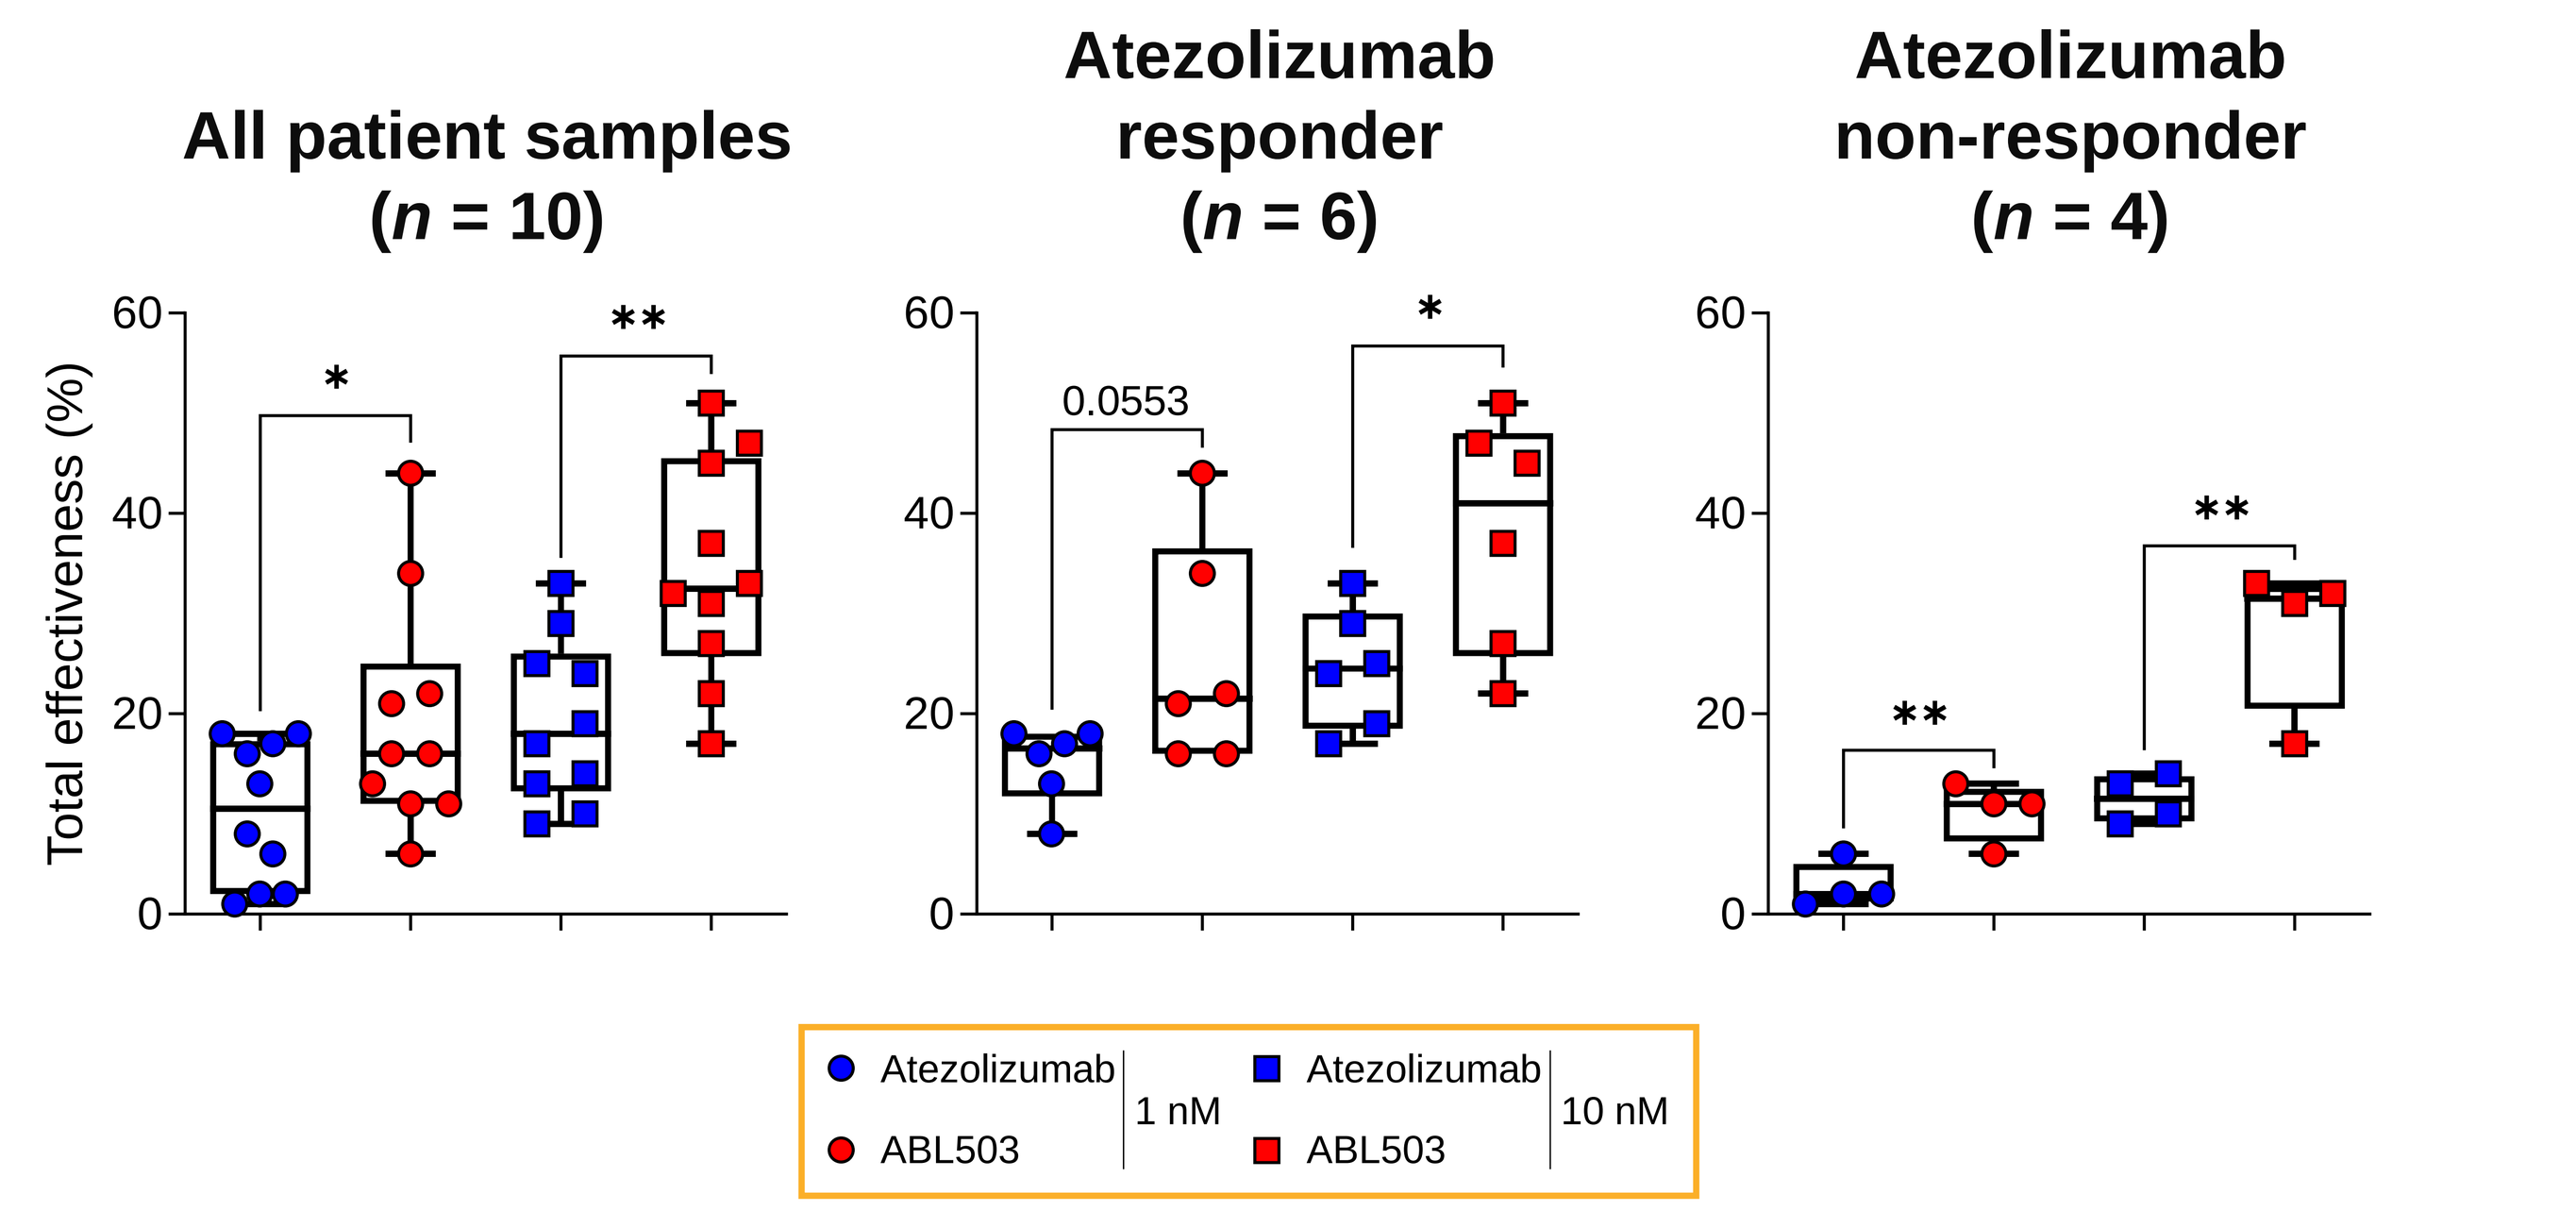
All patient samples (n = 10)
Atezolizumab responder (n = 6)
Atezolizumab non-responder (n = 4)
Total effectiveness (%)
Atezolizumab
ABL503
1 nM
10 nM

- Cancer organoids (n = 10) from lung cancer patients co-cultured with autologous PBMCs for 3 days in the presence of Atezolizumab or ABL503 (TJ-L14B) (1 or 10 nM).
- ABL503 (TJ-L14B) showed enhanced tumor-killing activity over Atezolizumab, even in organoids from Atezolizumab non-responder.

Pharmacodynamic changes by ABL503

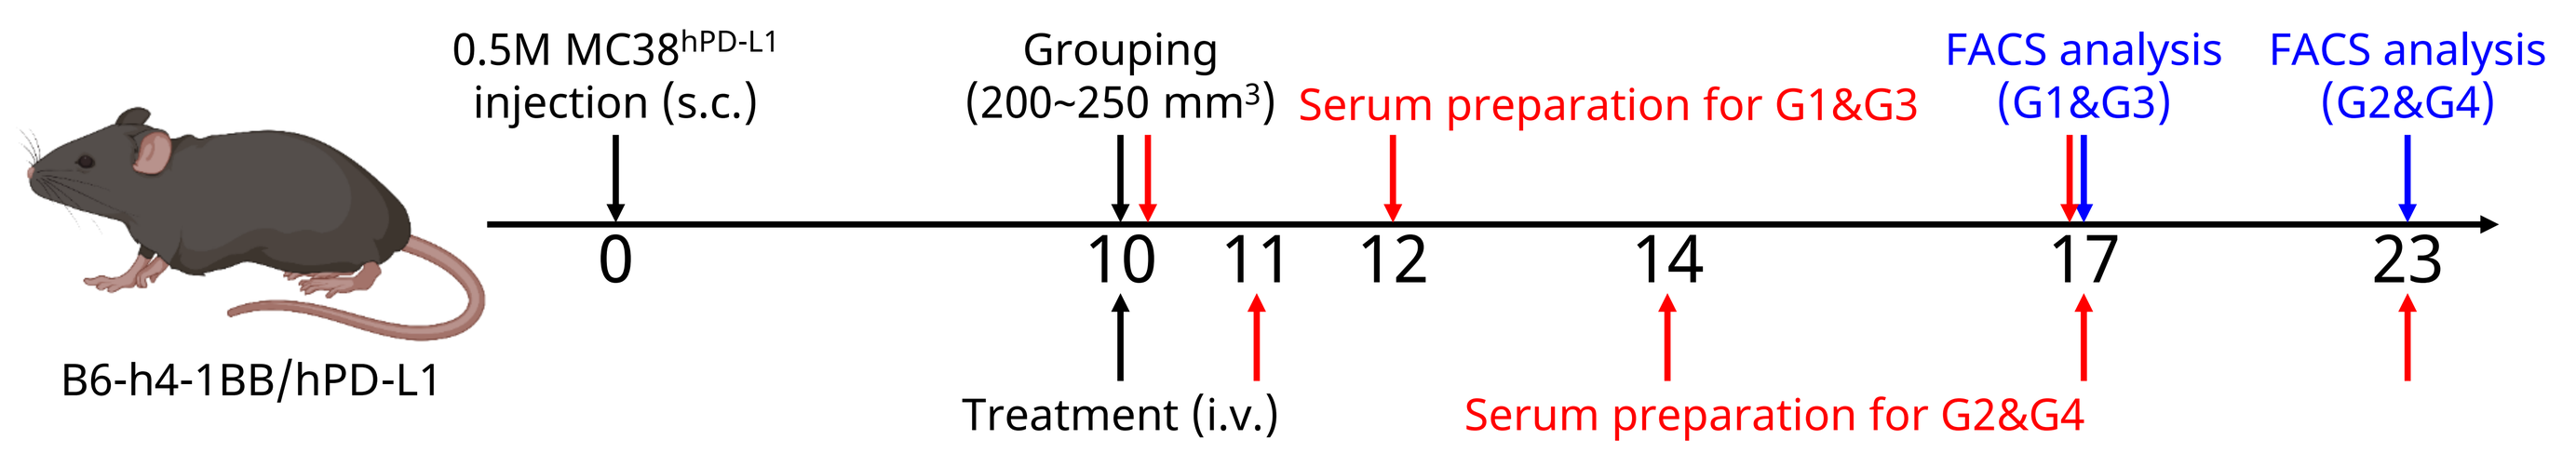
0.5M MC38hPD-L1 injection (s.c.)
Grouping (200~250 mm³)
Serum preparation for G1&G3
FACS analysis (G1&G3)
FACS analysis (G2&G4)
B6-h4-1BB/hPD-L1
Treatment (i.v.)
Serum preparation for G2&G4

Group	Treatment	Dose (mg/kg)	Group size	Analysis
G1	hIgG1	7.5, i.v.	n = 5/group	Cytokine, Immune phenotyping (Blood & TIL)
G2				s4-1BB, Immune phenotyping (Blood & TIL)
G3	ABL503	10, i.v.		Cytokine, Immune phenotyping (Blood & TIL)
G4				s4-1BB, Immune phenotyping (Blood & TIL)

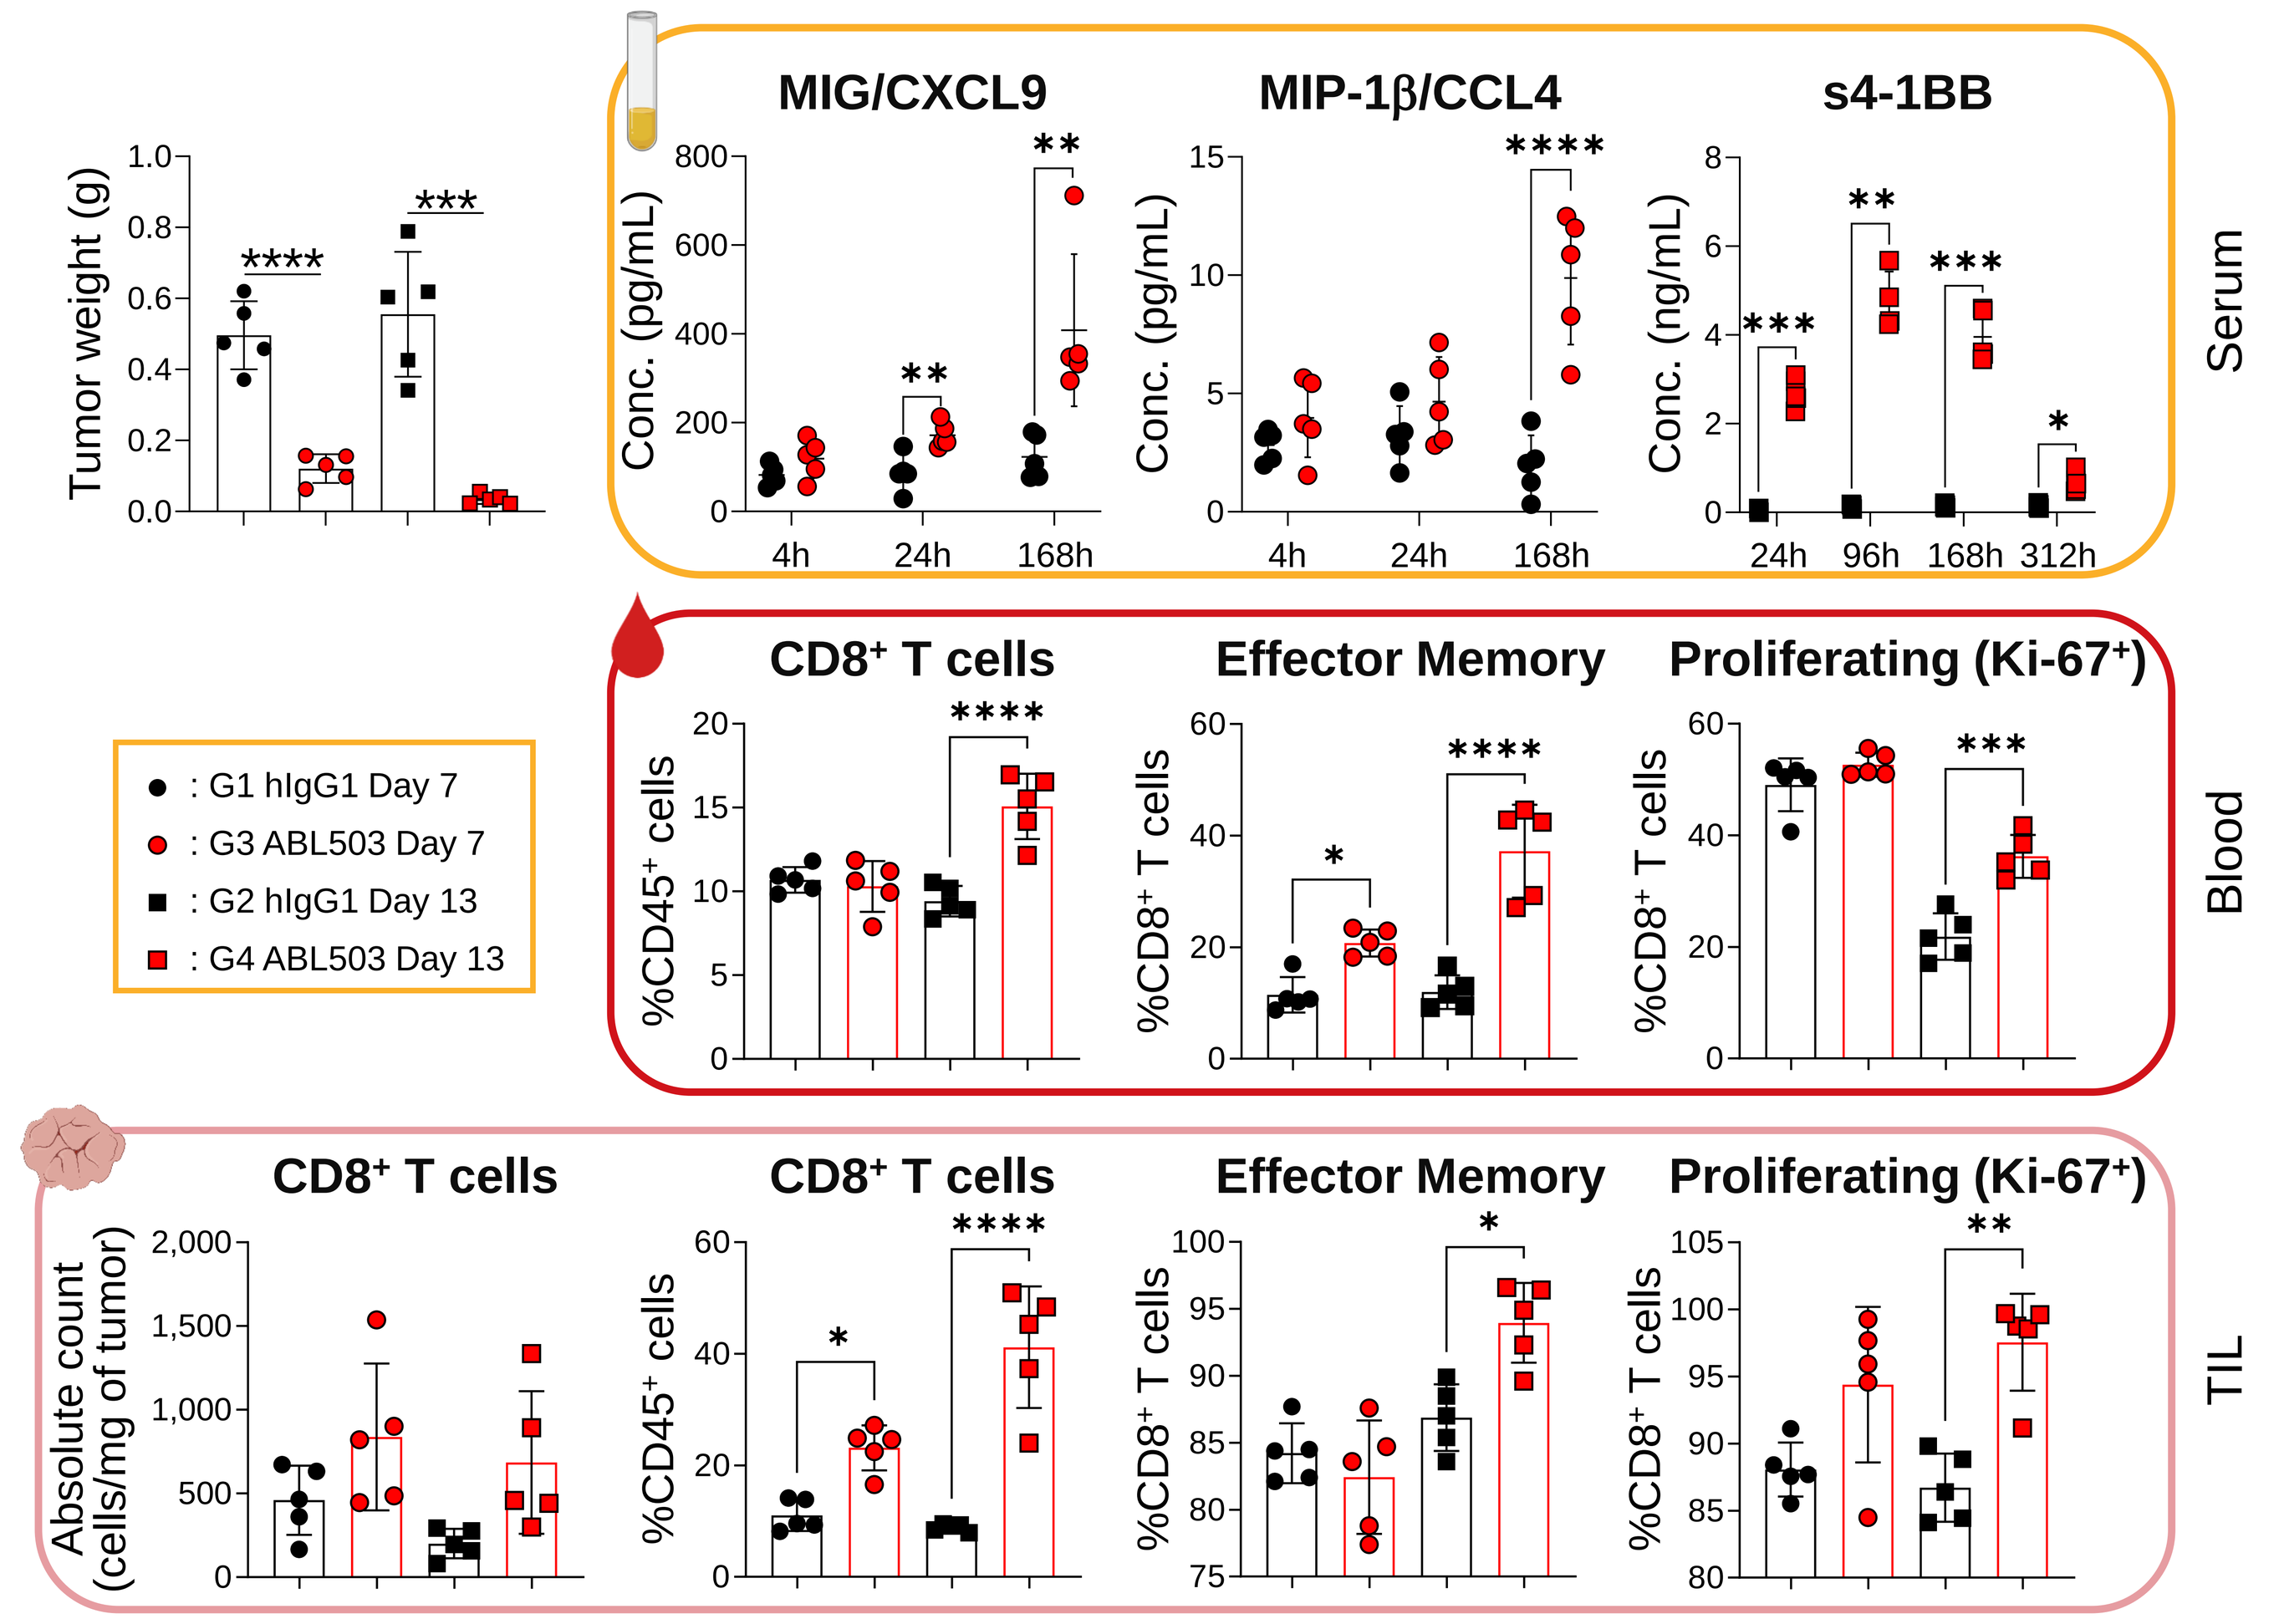
Tumor weight (g)
MIG/CXCL9
MIP-1β/CCL4
s4-1BB
Serum
CD8⁺ T cells
Effector Memory
Proliferating (Ki-67⁺)
Blood
TIL
: G1 hIgG1 Day 7
: G3 ABL503 Day 7
: G2 hIgG1 Day 13
: G4 ABL503 Day 13

- Pharmacodynamics changes were assessed in peripheral blood and tumor from MC38^{hPD-L1}-bearing B6-hPD-L1/h4-1BB mice at 7 and 13 days after a single treatment of ABL503 (TJ-L14B).
- ABL503 (TJ-L14B) showed anti-tumor activity accompanied by an increase of MIG/CXCL9, MIP-1β/CCL4, and soluble 4-1BB (s4-1BB) in the serum.
- ABL503 (TJ-L14B) increased frequency of CD8⁺ T cells both in peripheral blood and tumor with enhanced proliferation and increased effector memory population.

Summary

- ✓ ABL503 (TJ-L14B) shows PD-L1-dependent 4-1BB agonistic activity, which is expected to minimize peripheral toxicity as shown in the cytokine release assay and requires PD-L1 on both tumor and immune cells for its optimal activity.
- ✓ ABL503 (TJ-L14B) shows anti-tumor efficacy across the hPD-L1 levels. Especially, ABL503 (TJ-L14B) shows superior anti-tumor efficacy over Atezolizumab in the tumor model with low hPD-L1 expression.
- ✓ ABL503 (TJ-L14B) increases 4-1BB⁺ cells and CD8⁺ T cells in the TME, accompanied by proliferation and phenotypic changes to effector memory type. These changes are also reflected in blood.
- ✓ ABL503 (TJ-L14B) increases MIG/CXCL9, MIP-1b/CCL4, and s4-1BB in the serum, which may be used for PD markers in clinical trials.
- ✓ ABL503 (TJ-L14B) exhibits enhanced tumor-killing activity over Atezolizumab in patient-derived lung cancer organoid system.
- ✓ ABL503 (TJ-L14B) is currently undergoing a phase 1 clinical trial in the U.S. (NCT04762641).

SITC 2021 Annual Meeting

* Contact Information: Jaeho Jung (jaeho.jung@ablbio.com)